AGMB-129, an investigational ALK5 inhibitor for the treatment of Fibrostenosing Crohn's Disease (FSCD), shows gastrointestinal (GI) restricted pharmacokinetics (PK) and a favorable safety profile in healthy subjects

Andrea Sáez-Borderías[1], Tim Van Kaem[2], Joan Albertí[3], Pieter-Jan Stiers[2], Caroline Sabadie[2], Rolf van Heeswijk[2], Tony Senso[4], Begoña Pampín[1], Ramon Bosser[4], Philippe Wiesel[5]

[1]Agomab Spain S.L.U, A Coruña, Spain, [2]Agomab Therapeutics NV, Antwerp, Belgium, [3]DMPK Consultant, Spain, [4]formerly Agomab Spain S.L.U, A Coruña, Spain, [5]Agomab US, Inc, Cambridge, MA, USA

AGOMAB

Background

AGMB-129 is an oral GI-restricted small molecule inhibitor of Activin Receptor-Like Kinase 5 (ALK5) intended for the treatment of Fibrostenosing Crohn's Disease (FSCD). It potently blocks signaling of the key pro-fibrotic cytokine Transforming Growth Factor-β (TGFβ) and is designed to avoid clinically relevant systemic exposure via high first-pass metabolism to overcome the severe toxicities reported for prior systemic ALK5 inhibitors.

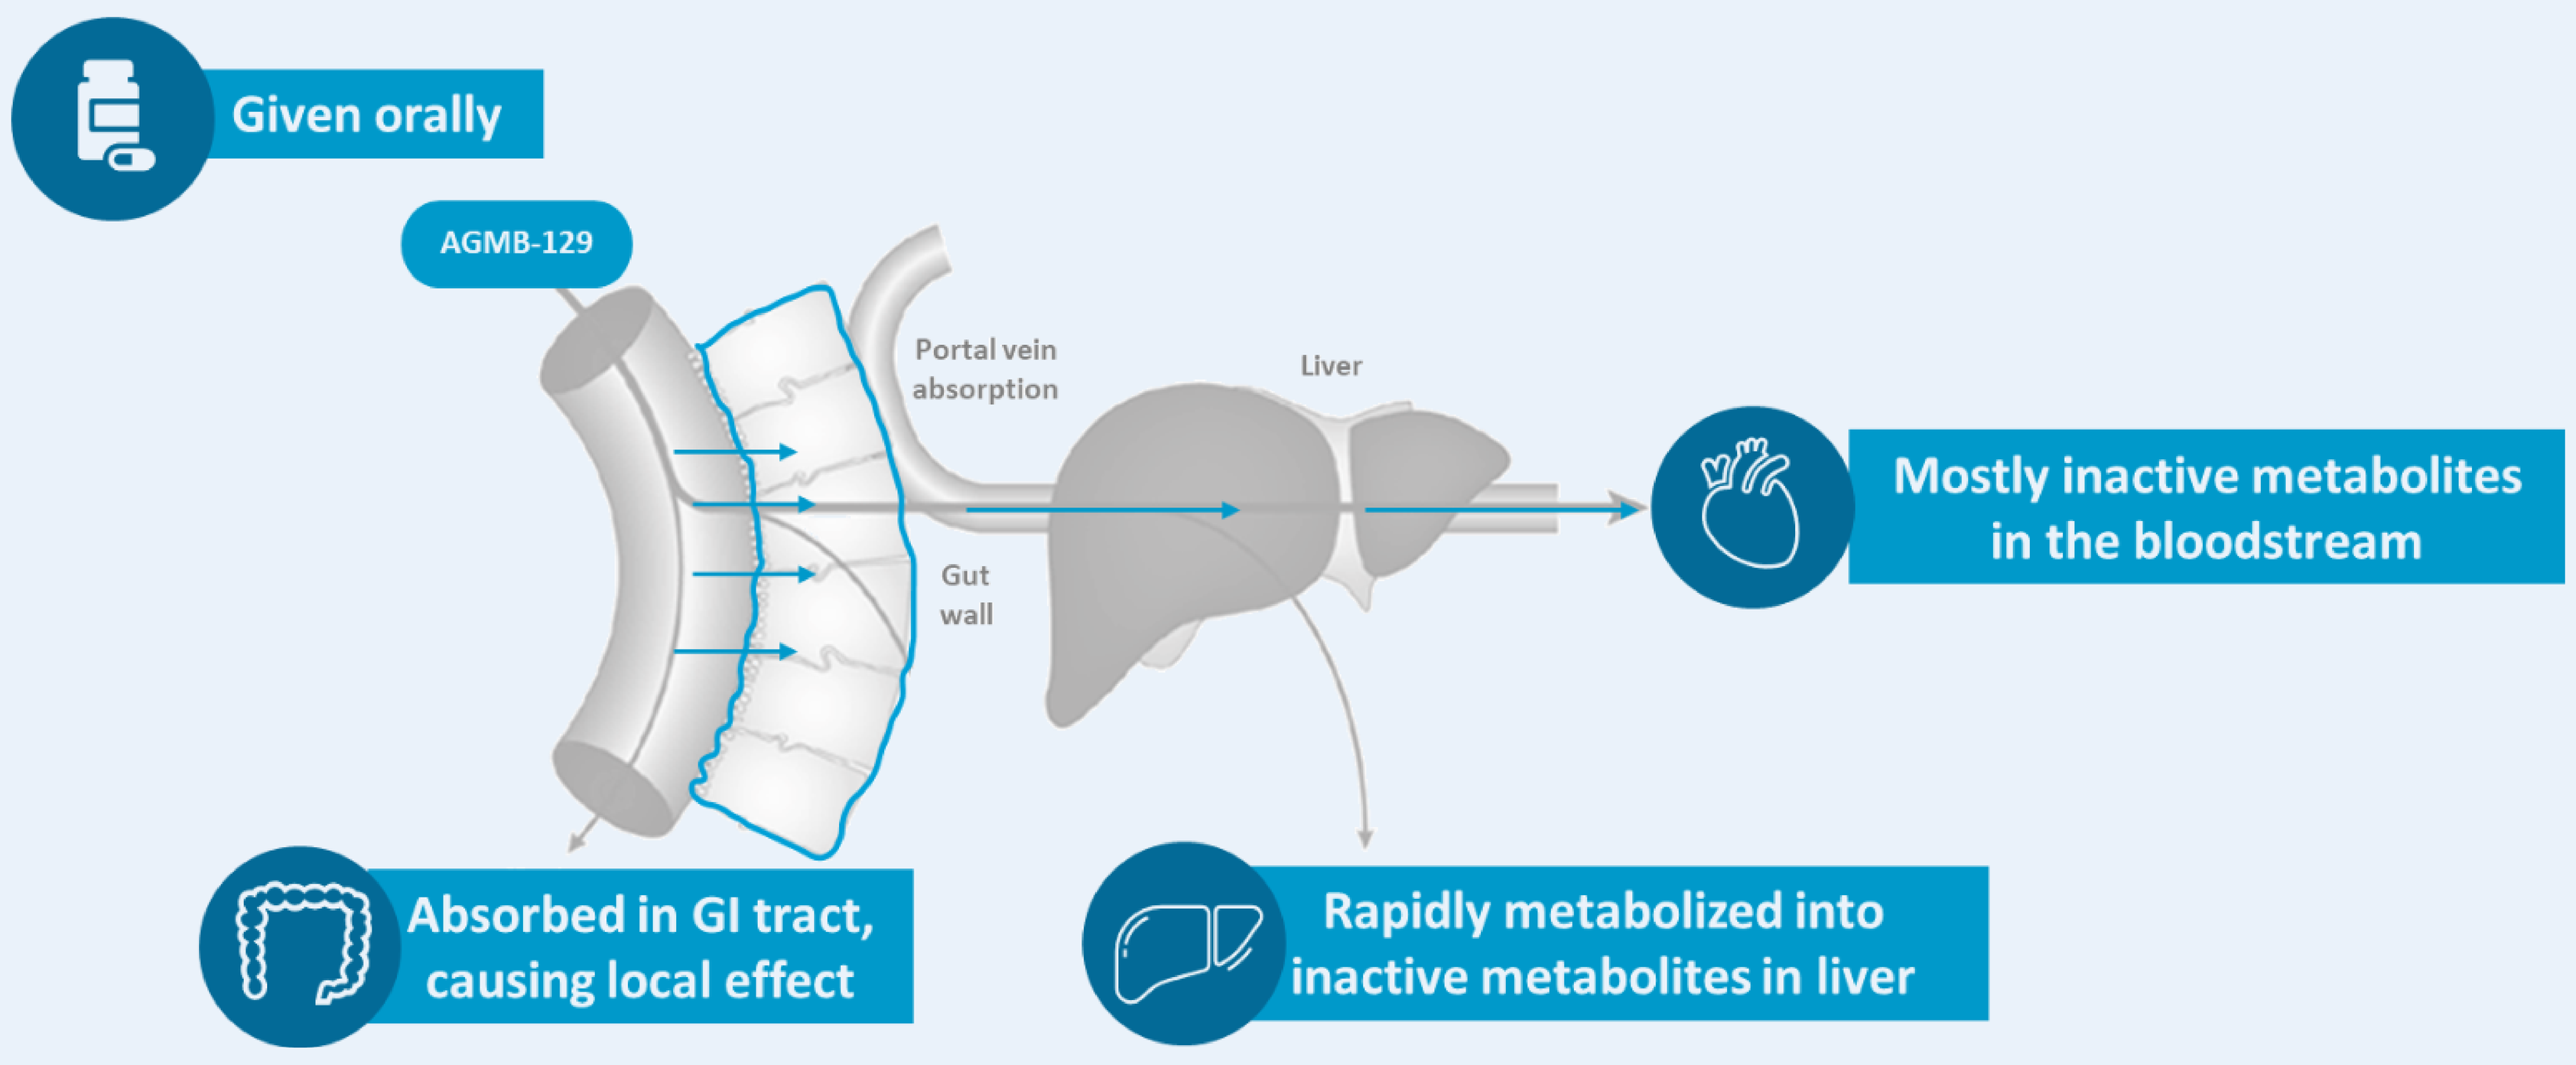
Given orally
AGMB-129
Portal vein absorption
Liver
Gut wall
Mostly inactive metabolites in the bloodstream
Absorbed in GI tract, causing local effect
Rapidly metabolized into inactive metabolites in liver

This study evaluated the safety, systemic and ileal PK of single- and multiple-doses AGMB-129 in healthy subjects.

Clinical study design

This was a randomized, double-blind, placebo-controlled, single and multiple ascending dose study (SAD/MAD) in healthy subjects. The effect of food on PK was evaluated in a cross-over fashion.

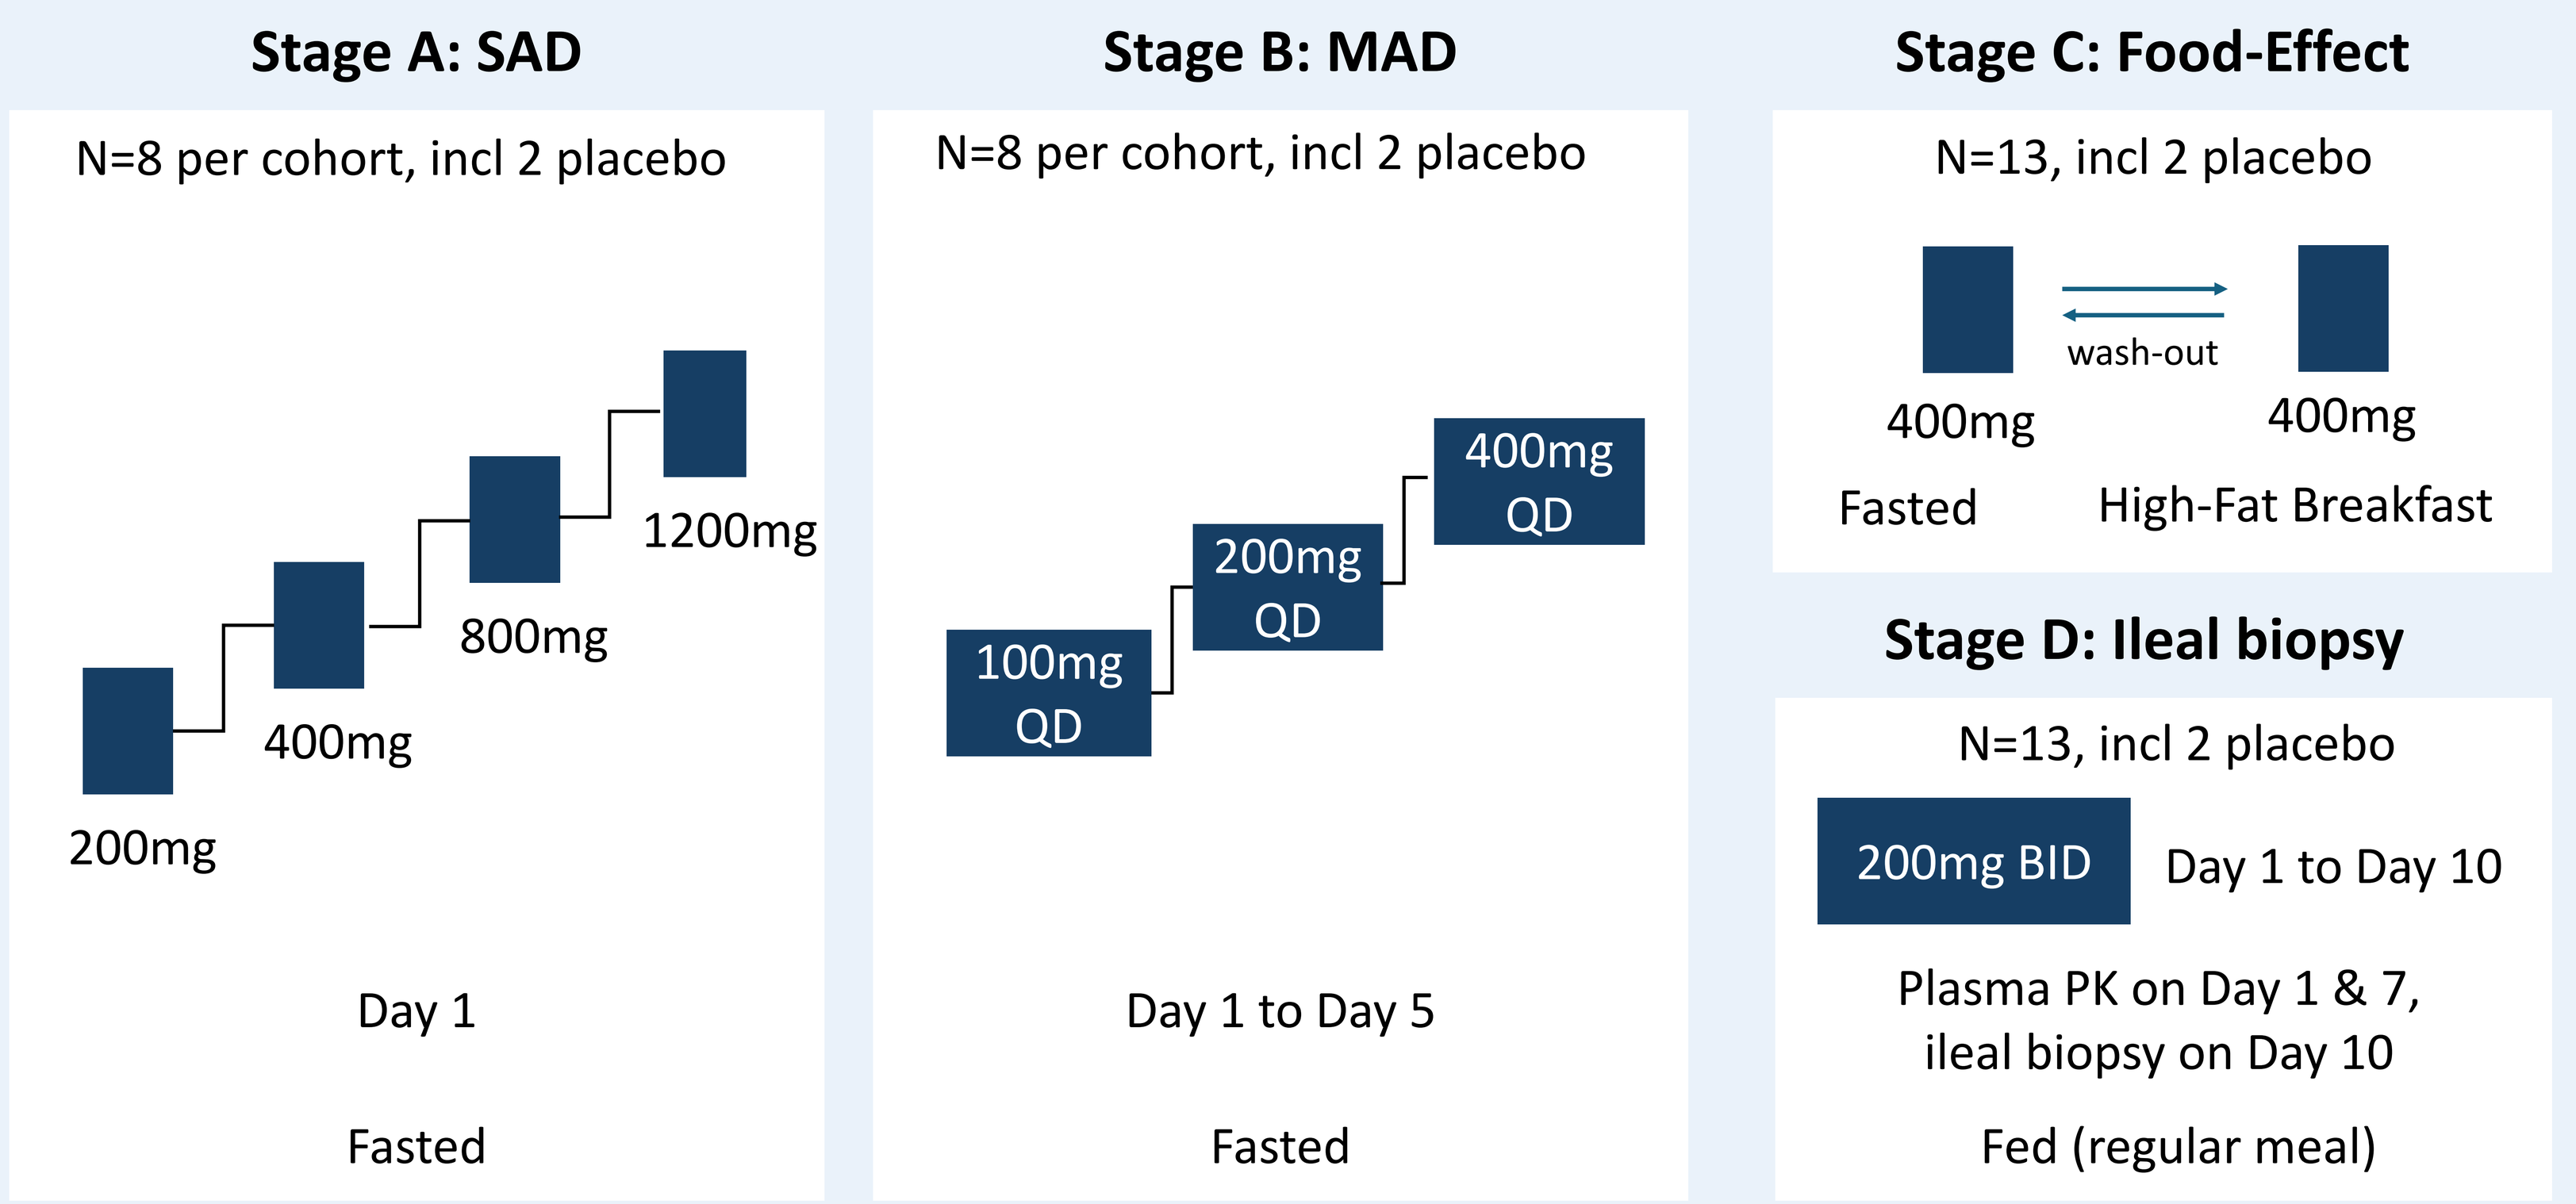
Stage A: SAD
N=8 per cohort, incl 2 placebo
200mg
400mg
800mg
1200mg
Day 1
Fasted
Stage B: MAD
N=8 per cohort, incl 2 placebo
100mg QD
200mg QD
400mg QD
Day 1 to Day 5
Fasted
Stage C: Food-Effect
N=13, incl 2 placebo
wash-out
400mg
Fasted
400mg
High-Fat Breakfast
Stage D: Ileal biopsy
N=13, incl 2 placebo
200mg BID
Day 1 to Day 10
Plasma PK on Day 1 & 7, ileal biopsy on Day 10
Fed (regular meal)

Results – Population & Safety

AGMB-129 was safe and well tolerated (Table 2), without dose-limiting toxicities, deaths, serious adverse events, or clinically-relevant abnormalities in laboratory results, vital signs or ECG parameters

Table 1. Study population (healthy subjects)

Stage	AGMB-129 (n)	Placebo (n)	Age (yr) Mean (range)	BMI (kg/m^2) Mean (range)	Male/ Female
A	24	8	27.6 (18-42)	24.3 (20-27)	32/0
B	18	6	32.5 (20-44)	23.9 (21-27)	15/9
C	11*	2	33.2 (24-43)	24.2 (21-27)	5/8
D	9	4*	31.9 (19-41)	24.8 (23-27)	8/5

*2 subjects discontinued due to viral infection (1 receiving AGMB-129 in Part C, 1 receiving placebo in Part D)

Table 2. Adverse Events

	Stage A, n (%)		Stage B, n (%)		Stage C, n (%)		Stage D, n (%)	
TEAE	AGMB-129	Placebo	AGMB-129	Placebo	AGMB-129	Placebo	AGMB-129	Placebo
Mild	5 (20.8)	2 (25.0)	6 (33.3)	2 (33.3)	6 (54.5)	0	1 (11.1)	0
Moderate	0	0	2 (11.1)	0	3 (27.3)	1 (50.0)	1 (11.1)	1 (25.0)
Severe	0	0	0	0	0	0	0	0
Related	4 (16.7)	2 (25.0)	8 (44.4)	2 (33.3)	7 (63.6)	1 (50.0)	1 (11.1)	0
Serious	0	0	0	0	0	0	0	0

TEAE = Treatment Emergent Adverse Event, n = number of subjects with an adverse event (AE)

Results - Pharmacokinetics

Stage A (SAD): After oral dosing, AGMB-129 was rapidly absorbed, followed by a rapid decline in plasma concentration to <1 ng/mL within 4 to 8h post-dose resulting in low systemic exposure at all doses (Figure 1). MET-158 (inactive against ALK5) was the main circulating metabolite. AGMB-129 C_{max} and AUC increased close to proportional to the dose (Table 3), based on a power analysis (power coefficient β for C_{max} and AUC were 1.13 and 1.18, resp., 95% CI: 0.4-1.9 for both parameters).

Figure 1. Geometric Mean (GeoSD) plasma concentrations of AGMB-129 after single-dose administration without food

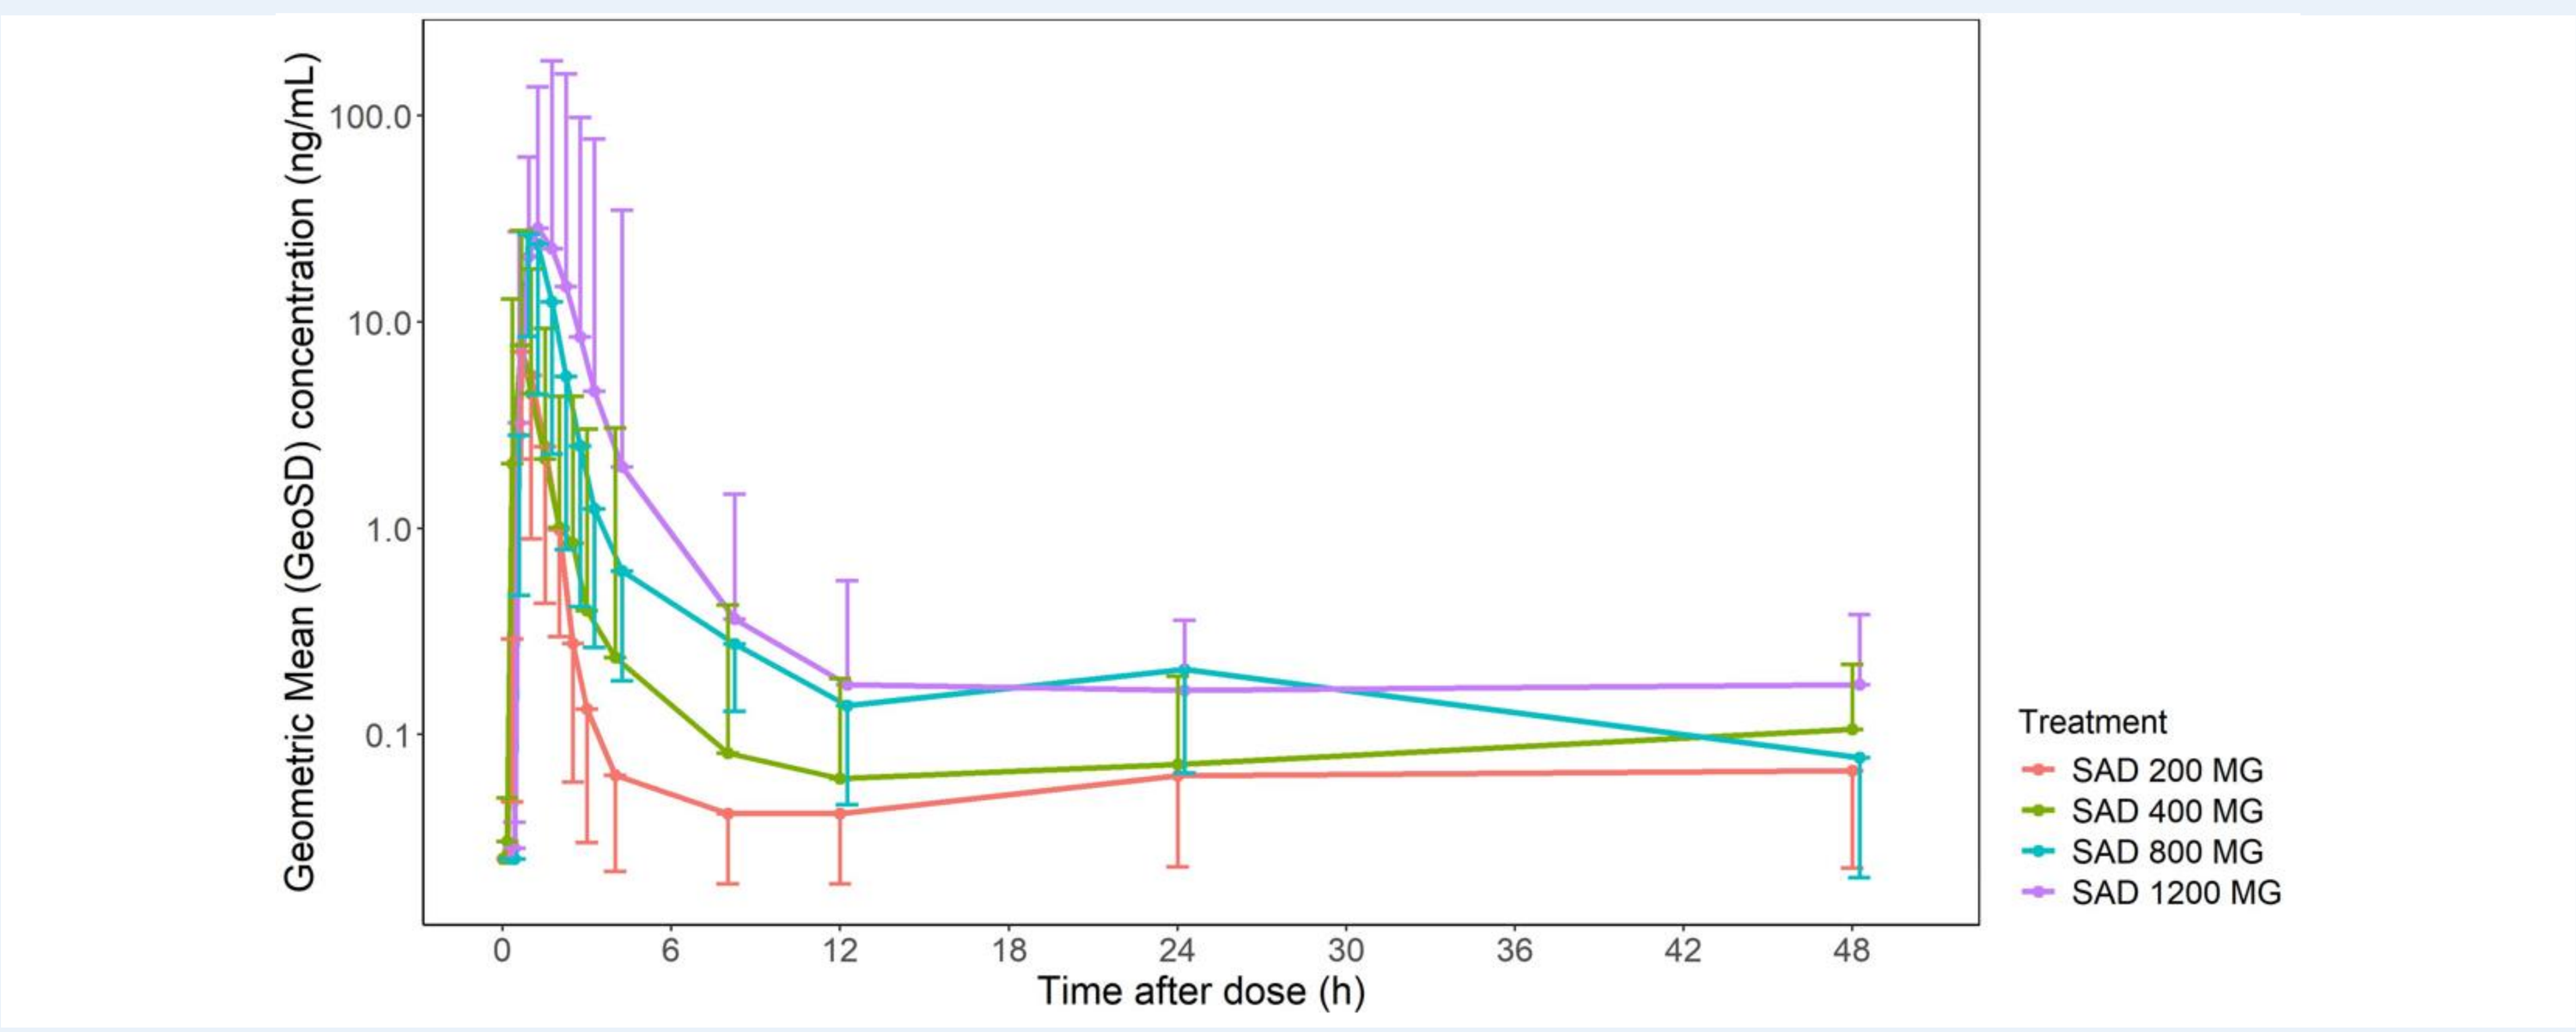
Geometric Mean (GeoSD) concentration (ng/mL)
100.0
10.0
1.0
0.1
0
6
12
18
24
30
36
42
48
Time after dose (h)
Treatment
SAD 200 MG
SAD 400 MG
SAD 800 MG
SAD 1200 MG

Table 3. Geometric Mean (%gCV) C_{max} and AUC after single-dose administration

Part A	AGMB-129		MET-158	
Single-dose	C_{max} (ng/mL)	AUC_{0-t} (h.ng/mL)	C_{max} (ng/mL)	AUC_{0-t} (h.ng/mL)
200mg	11 (199)	14 (85)	683 (134)	3149 (79)
400mg	10 (252)	14 (276)	1000 (81)	4011 (80)
800mg	48 (154)	57 (129)	1466 (159)	5150 (143)
1200mg	63 (198)	100 (293)	2868 (57)	11931 (100)

AUC0-t = area under the curve from time 0 extrapolated to the last measurable concentration; Cmax = max plasma concentration; gCV% = geometric % coefficient of variation

Stage B (MAD): After QD dosing for 5 days, AGMB-129 accumulation based on the AUC was limited (<2-fold). AGMB-129 and MET-158 AUC increased close to dose-proportional, and systemic exposure to AGMB-129 was low across the dose range (Figure 2 and Table 4).

Figure 2. Geometric Mean AGMB-129 and MET-158 AUC24h on Day 5 versus Dose

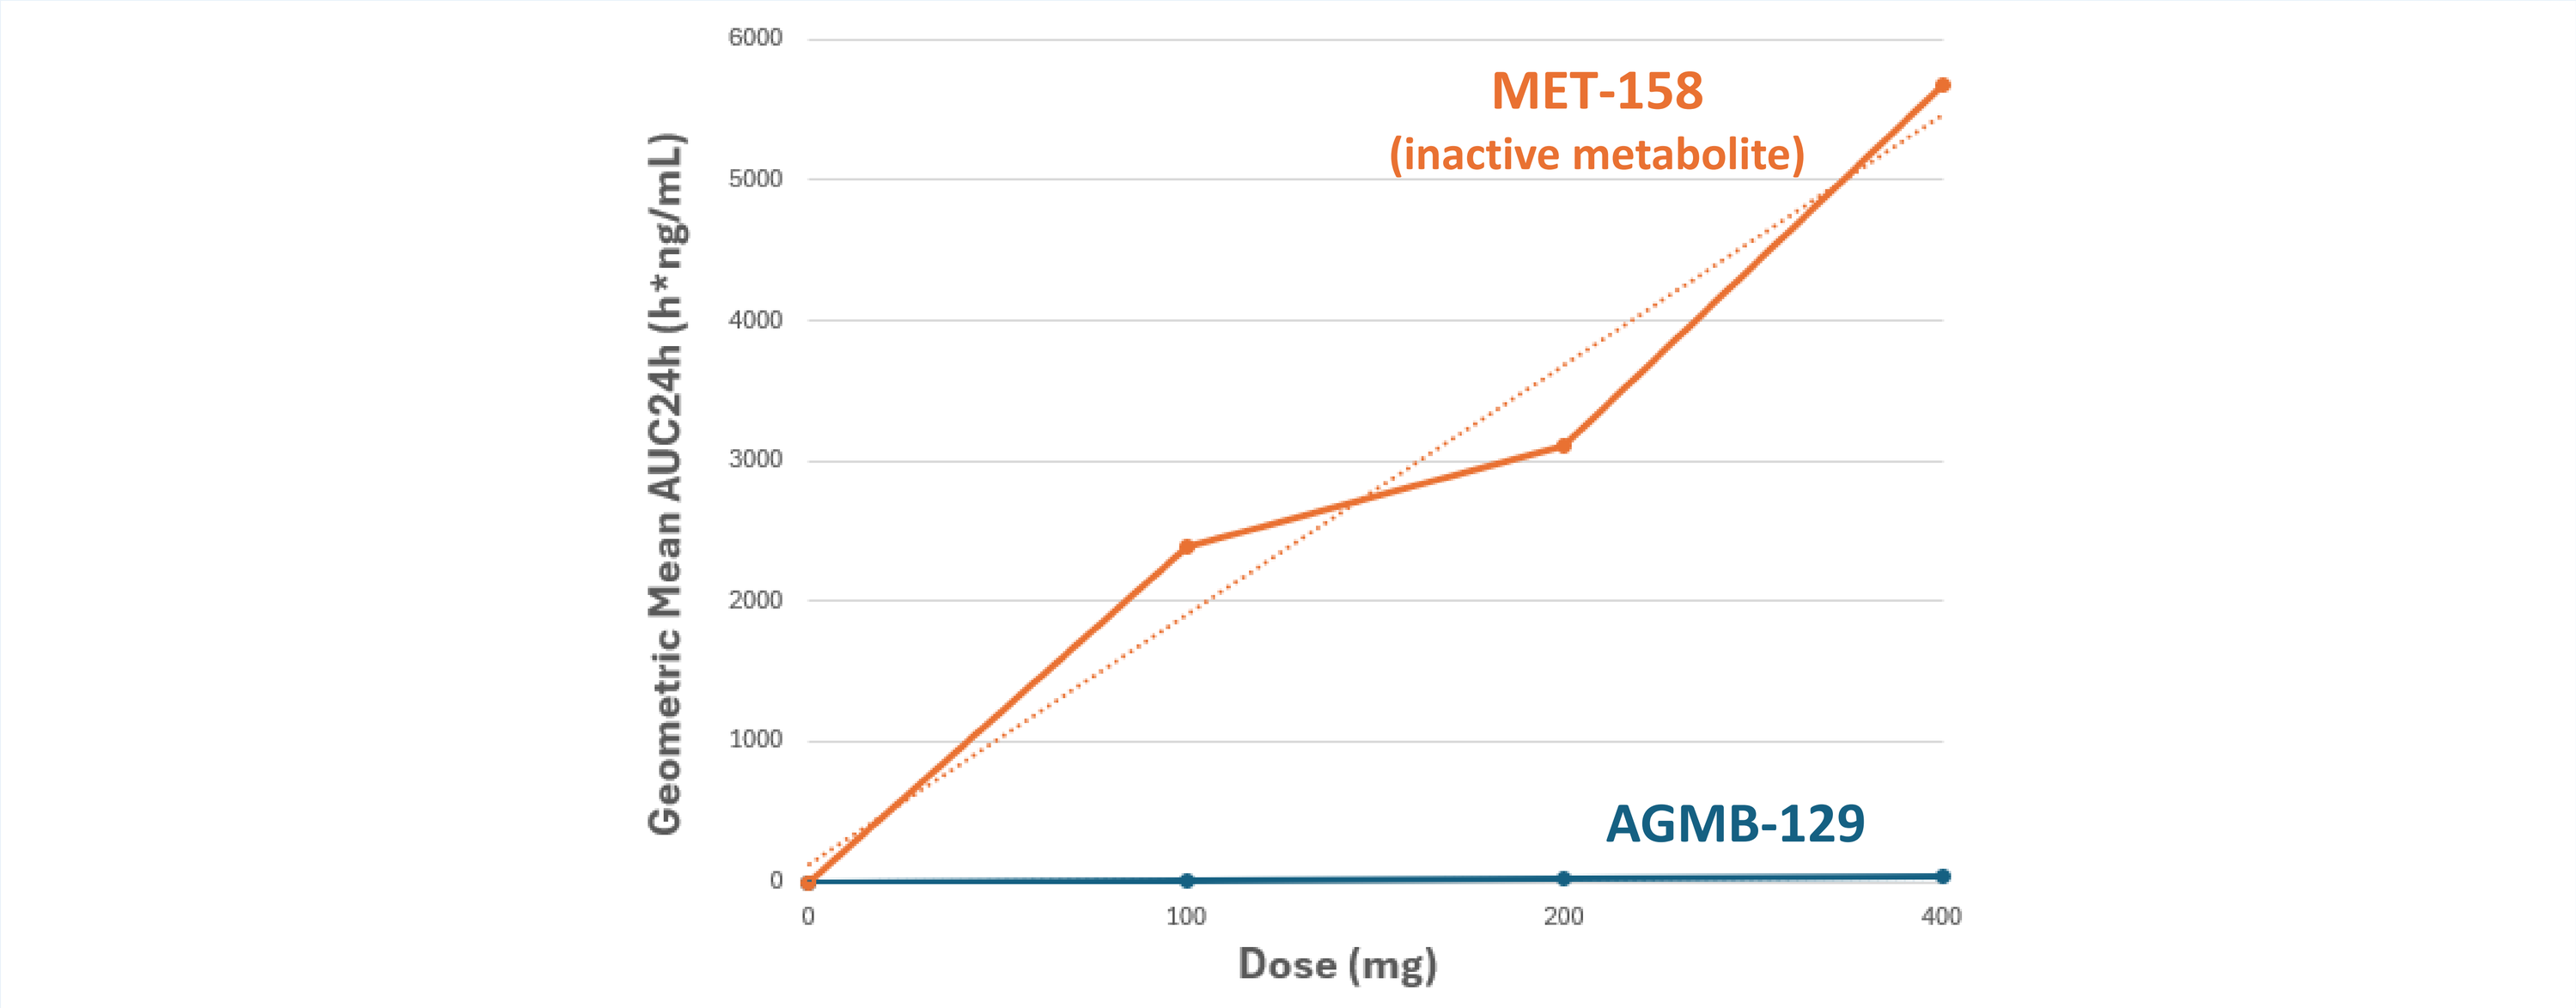
Geometric Mean AUC24h (h*ng/mL)
6000
5000
4000
3000
2000
1000
0
MET-158
(inactive metabolite)
AGMB-129
0
100
200
400
Dose (mg)

Table 4. Geometric Mean (%gCV) C_{max} and AUC after 5 days of QD dosing

Stage B	AGMB-129		MET-158	
QDx5	C_{max} (ng/mL)	AUC_{24h} (h.ng/mL)	C_{max} (ng/mL)	$AUC24_h$ (h.ng/mL)
100mg	11 (52)	9 (44)	857 (18)	2380 (27)
200mg	18 (125)	21 (95)	888 (113)	3102 (88)
400mg	24 (556)	36 (113)	1327 (362)	5683 (135)

AUC24h = area under the curve during the dosing interval; Cmax = max plasma concentration; gCV% = geometric % coefficient of variation

Stage C (Food-Effect): After intake with food, AGMB-129 T_{max} was delayed by about 2h, C_{max} and AUC increased by 1.3- and 2-fold, respectively, and intersubject variability was reduced compared to fasted intake.

Stage D (Biopsy cohort): After AGMB-129 200 mg BID (fed), there was limited accumulation from Day 1 to Day 7 for both AGMB-129 and MET-158. The gMean AGMB-129 total **concentration in ileal biopsies** (20 mg tissue) at 2h post-dose on Day 10 was **4,243 ng/mL** (range 666 ng/mL to 57 µg/mL, n=8, excl. 1 high outlier).

Conclusion

After oral dosing, low systemic and high ileal exposure to AGMB-129 was observed. This confirms the GI-restricted profile of AGMB-129 with systemic exposure more than 100-fold lower than previously observed for systemic ALK5 inhibitors [Rodón 2015, Jung 2020]. AGMB-129 was safe and well-tolerated at all dosages and is the first ALK5 inhibitor suitable for continuous daily dosing in humans. The STENOVA Ph2a study of AGMB-129 up to 200 mg BID in symptomatic FSCD is ongoing (NCT05843578).

European Crohn's and Colitis Organisation
This poster was presented at the annual ECCO Congress.

ECCO 2025, February 19-22, Berlin, Germany, Poster P0334

Disclosures: AS, TVK, PJS, CS, RVH, BP, PW are current employees of Agomab, TS and RB are former employees of Agomab

Agomab Therapeutics NV
Posthoflei 1/6, 2600 Antwerp, Belgium
www.Agomab.com

Andrea Sáez-Borderías
andrea.saez@agomab.com